Markers of HIV reservoir size in infected children on long-term viral control 0810

Stefano Rinaldi[1], Suresh Pallikkuth[1], Lesley de Armas[1], Rajendra Pahwa[1], Sara Dominguez[2], Nicola Cotugno[3], Pablo Rojo Conejo[2], Eleni Nastouli[4], Kathleen Gartner[4], Nigel Klein[4], Caroline Foster[5], Anita De Rossi[6], Carlo Giaquinto[7], Paolo Rossi[2], Paolo Palma[3], Savita Pahwa[1] and EPIICAL Consortium

[1]Department of Microbiology and Immunology, University of Miami Miller School of Medicine, Miami, FL, USA [2]Hospital Universitario 12 de Octubre, Madrid, Spain, [3]Bambino Gesu Children's Hospital, Rome, Italy, [4]UCL Great Ormond Street Institute of Child Health, London, UK, [5]Imperial College Healthcare NHS Trust, London, UK, [6]Istituto Oncologico Veneto IOV-IRCCS, Padova, Italy, [7]University of Padova, Padova, Italy

Abstract

Introduction: Curative strategies for HIV will need to eliminate the replication competent latent reservoir. CD4 T cells expressing Immune Checkpoint molecules (ICP) have been shown to preferentially harbor latent, replication-competent HIV. T follicular helper (Tfh) cell subset of CD4 T cells are critical for B cell differentiation. Here a cohort of HIV vertically infected children and young adults under durable viral control (PHIV) were investigated for CD4 ICP, immune activation (IA) markers and function in relation to HIV reservoir size.

Methods: 40 PHIV (4-19yrs age) who started ART <2 years of life and had undetectable viremia (<50 HIV copies/ml) for the past 5 years, were enrolled in 7 European research centers. HIV-DNA copies per million peripheral blood mononuclear cells (PBMC) were measured by real-time PCR. Flow cytometry was used to investigate CD4 T cells for 1) co-expression of PD1 with IA (ICOS, CD38, Ki67 and HLA-DR) or ICP (TIGIT, LAG3, TIM3 and CTLA4) and 2) intracellular cytokine production (IL2, IFNg, TNFα, IL21) after stimulation with GAG peptides.

Results: We identified a T cell immune signature that associated with total HIV DNA based on markers of immune activation, exhaustion and functions. Interestingly, both the sPLS-DA and Poisson regression approaches identified PD-1 expressing CD4 cells and HIV responsive (CD40L+) CD4 T cell to be important to predict the size of viral reservoir. Using univariate correlation we also identify a correlation between PD-1 CD4 T cells and both CD4 and CD8 functions.

Conclusion: This study suggests that PD-1 expression on CD4 T cells is associated with dysfunctional T cell response to HIV antigens and supports the association of PD1 expression on CD4 T cells with size of viral reservoirs in vertically HIV infected children and young adults under long-term viral control.

Background

Curative strategies for HIV will need to eliminate the replication competent latent reservoir.

Immune Checkpoint Molecules (ICP), a group of proteins highly expressed on exhausted immune cells, are promising therapeutic targets for the elimination of the HIV latent reservoir, as CD4 T cells expressing ICP have been shown to preferentially harbor latent, replication-competent HIV. (Fromentin *et al*. Plos Pathog 2016).

T follicular helper (Tfh) cells, a subset of CD4 T cells critical for B cell differentiation, were shown to harbor HIV and to be functionally impaired in infected individuals (Pallikkuth *et al*. Plos Biology 2019).

Here a cohort of HIV vertically infected children and young adults under durable viral control were investigated for CD4 ICP, immune activation (IA) markers and GAG specific (GAGsp) CD4 and CD8 T cell function in relation to HIV reservoir size.

Study population

- 40 perinatally HIV infected young and old children (age range 4.4-19.1 years) were enrolled in the study
- All study participants had durable viral control (HIV RNA load, <50 copies/mL)
- Selection of the participants with high and low HIV-DNA was made based on data distribution. Participants with HIV-DNA in the 1st quartile (HIV DNA ≤ 12.3 copies/mL) were considered to have low HIV-DNA while participants in the 4th quartile (HIV DNA ≥ 112.7 copies/mL) were considered to have high HIV-DNA.

Demographic characteristics of study participants.

# individuals	40
Median Age at sampling (Range) Years	12.2 (4.4-19.1)
Median CD4 Absolute Count (Range) cells/uL	1515 (95-5166)
Median CD8 Absolute Count (Range) cells/uL	1406 (110-5582)
Median CD4/CD8 ratio (Range)	0.9 (0.3-2.9)
HIV-caDNA (Copies/10^6 PBMC)	48.3 (0-323.1)
Time of Treatment Initiation (Range) Months	2(0-17)

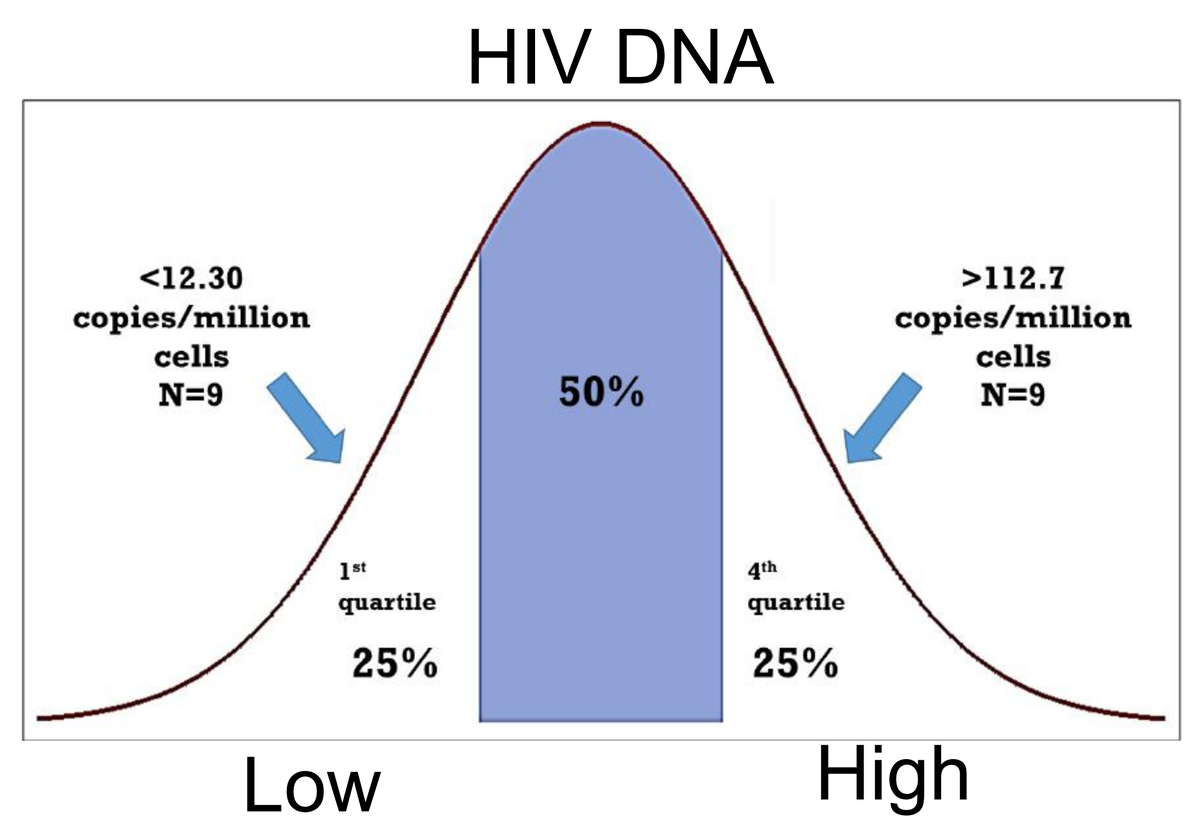
HIV DNA
<12.30 copies/million cells N=9
>112.7 copies/million cells N=9
50%
1st quartile
25%
4th quartile
25%
Low
High

Methods

Flow cytometry data sets generation and integration

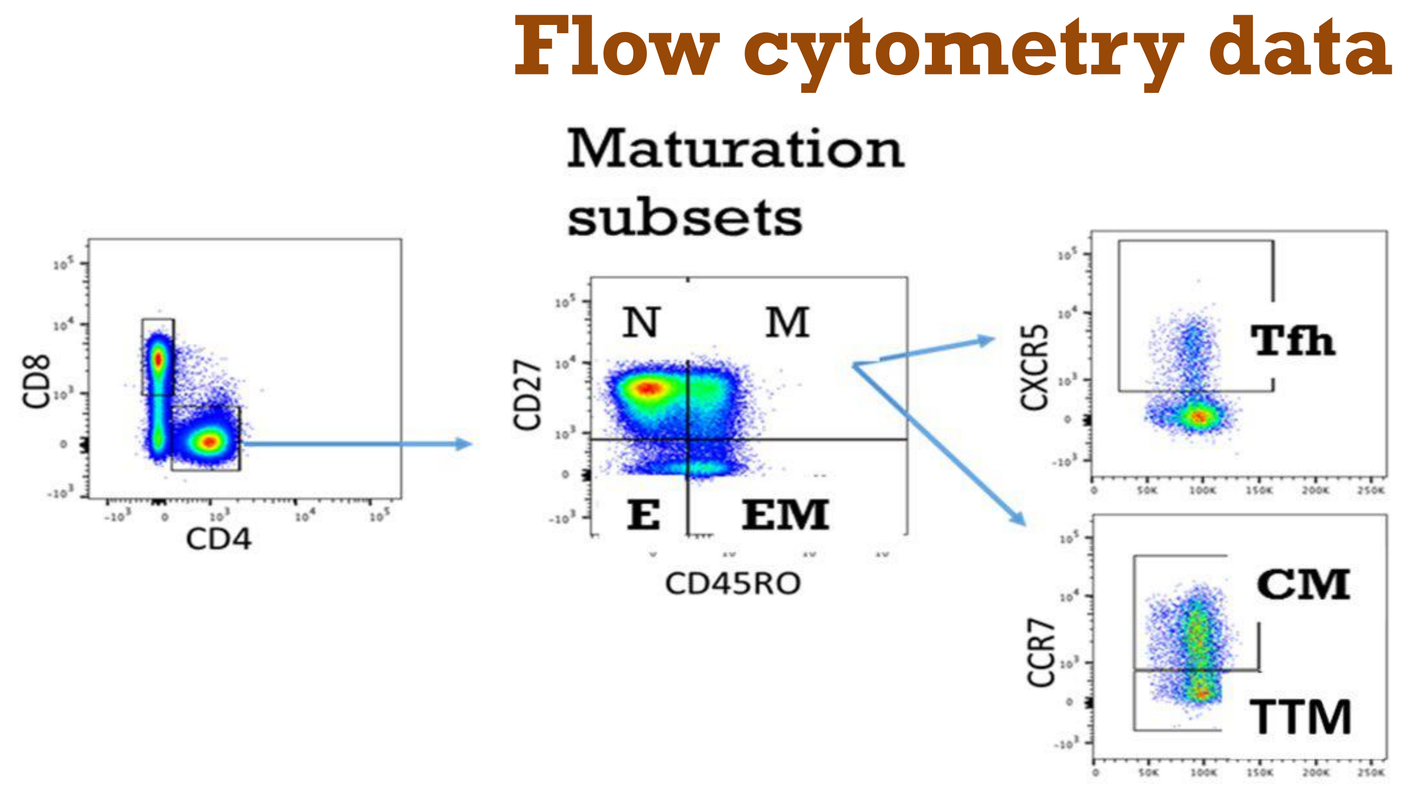
Maturation subsets
CD8
CD4
CD27
CD45RO
N
M
E
EM
CXCR5
Tfh
CCR7
CM
TTM

Legend:
N: Naïve
M: Memory
E: Effector
EM: Effector Memory
Tfh: T follicular Helper
CM: Central Memory
TTM: Transitional Memory

Immune Activation	Immune Checkpoint	T cell function
PD-1/ CD38/ HLA-DR/ ICOS/ Ki67	PD-1/ LAG3/ TIGIT3/ TIM3/ CTLA4	CD40L/ CD69 CD107a/ Perf/ GrzB IFNγ/ TNF/ IL2/ IL-21

Evaluated after stimulation with GAG PTE peptides

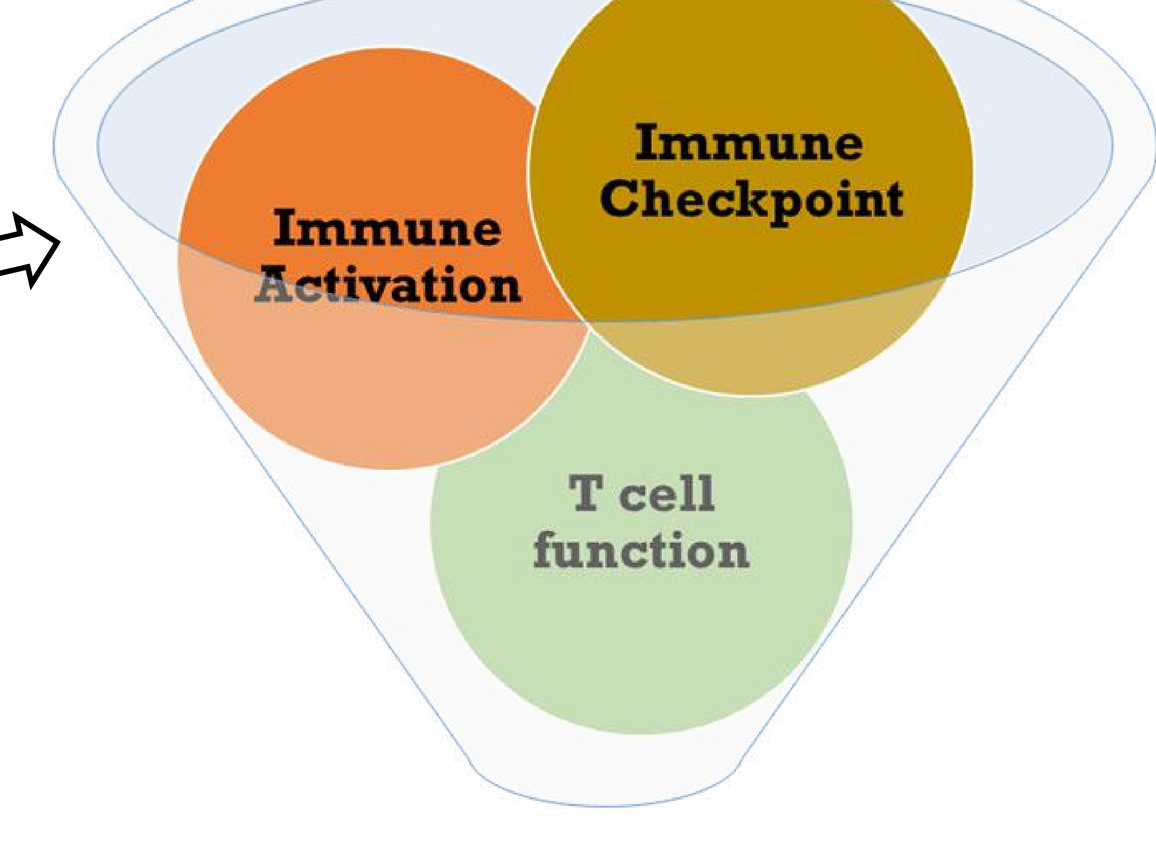
Immune Activation
Immune Checkpoint
T cell function

PD-1 EXPRESSION ON CD4 T CELLS IS ASSOCIATED WITH HIGHER HIV RESERVOIR AND POOR CD8 T CELL RESPONSE

Methods (contd.)

Statistical analysis

sPLS-DA, Penalized regression, multivariable Poisson regression and hierarchical clustering analyses were performed using R (R Core Team (2014). R: A language and environment for statistical computing. R Foundation for Statistical Computing, Vienna, Austria. URL http://www.R-project.org/).

Results

Fig 1. PD-1+ and CD40L+ T cells were identified as immunological predictors that could discriminate participants with high and low HIV DNA by sPLS-DA

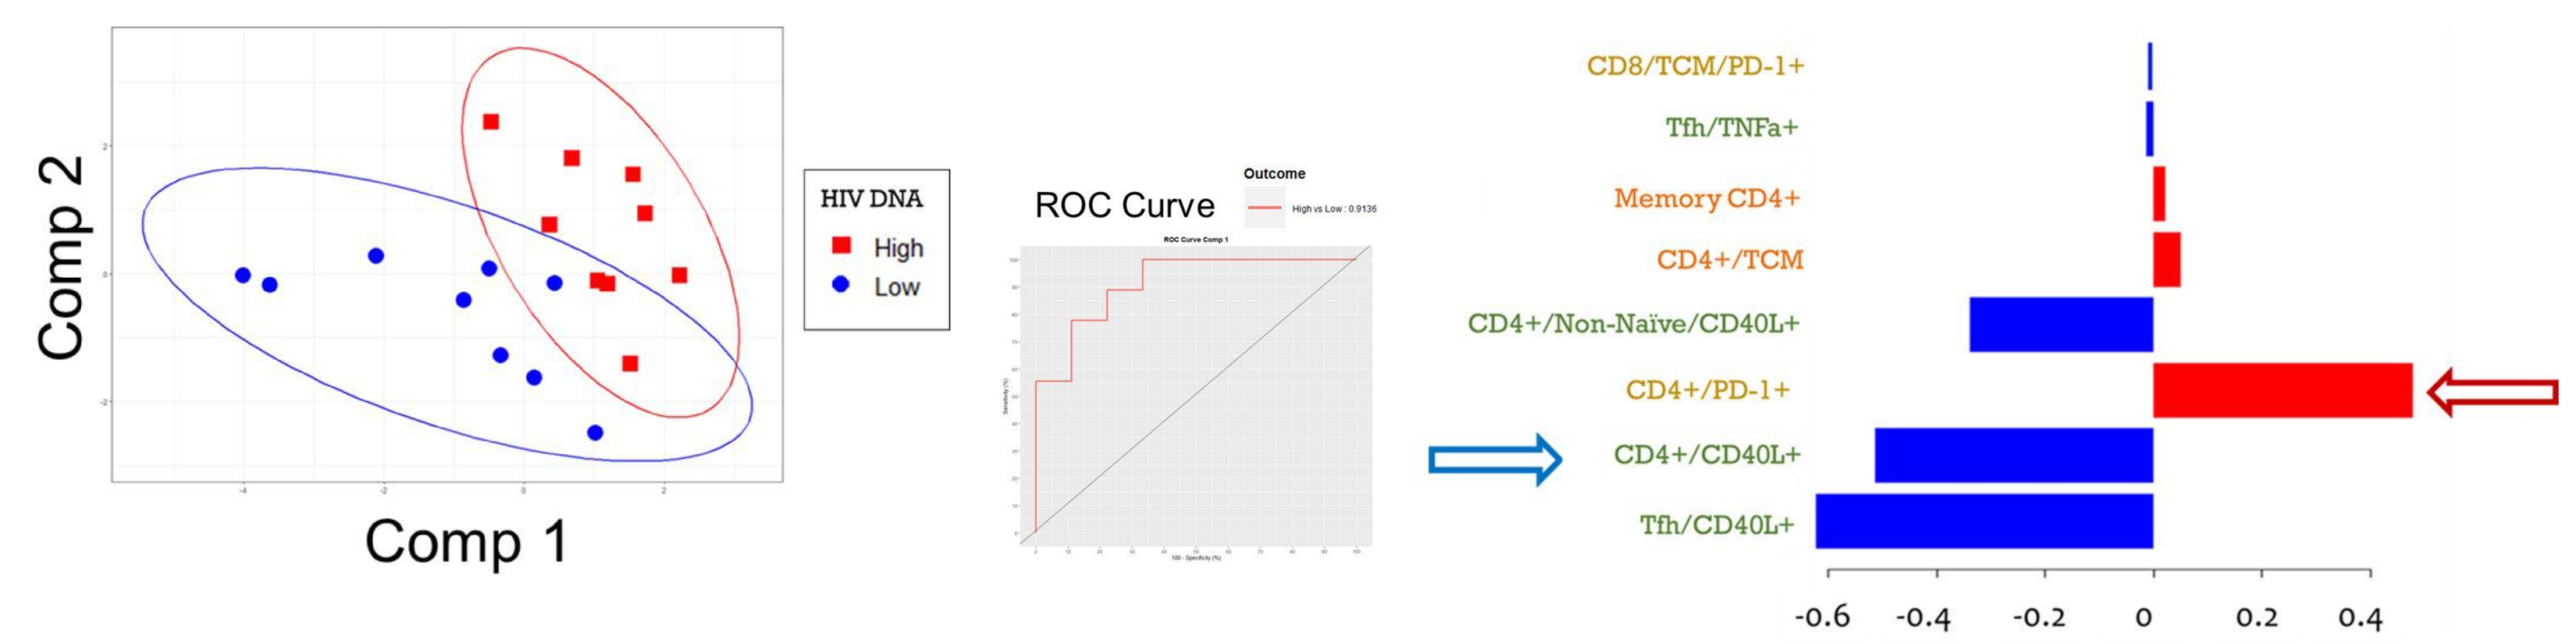
Comp 2
Comp 1
HIV DNA
High
Low
ROC Curve
Outcome
CD8/TCM/PD-1+
Tfh/TNFa+
Memory CD4+
CD4+/TCM
CD4+/Non-Naïve/CD40L+
CD4+/PD-1+
CD4+/CD40L+
Tfh/CD40L+
-0.6
-0.4
-0.2
0
0.2
0.4

Antigen responding CD4 T cells associates with low HIV-DNA
PD-1+ CD4 T cells associates with high HIV-DNA

Fig 2. PD-1+ and CD40L+ CD4 T cell associate with HIV-1 DNA reservoir, and interact with age at Treatment initiation (TI) using Poisson Regression

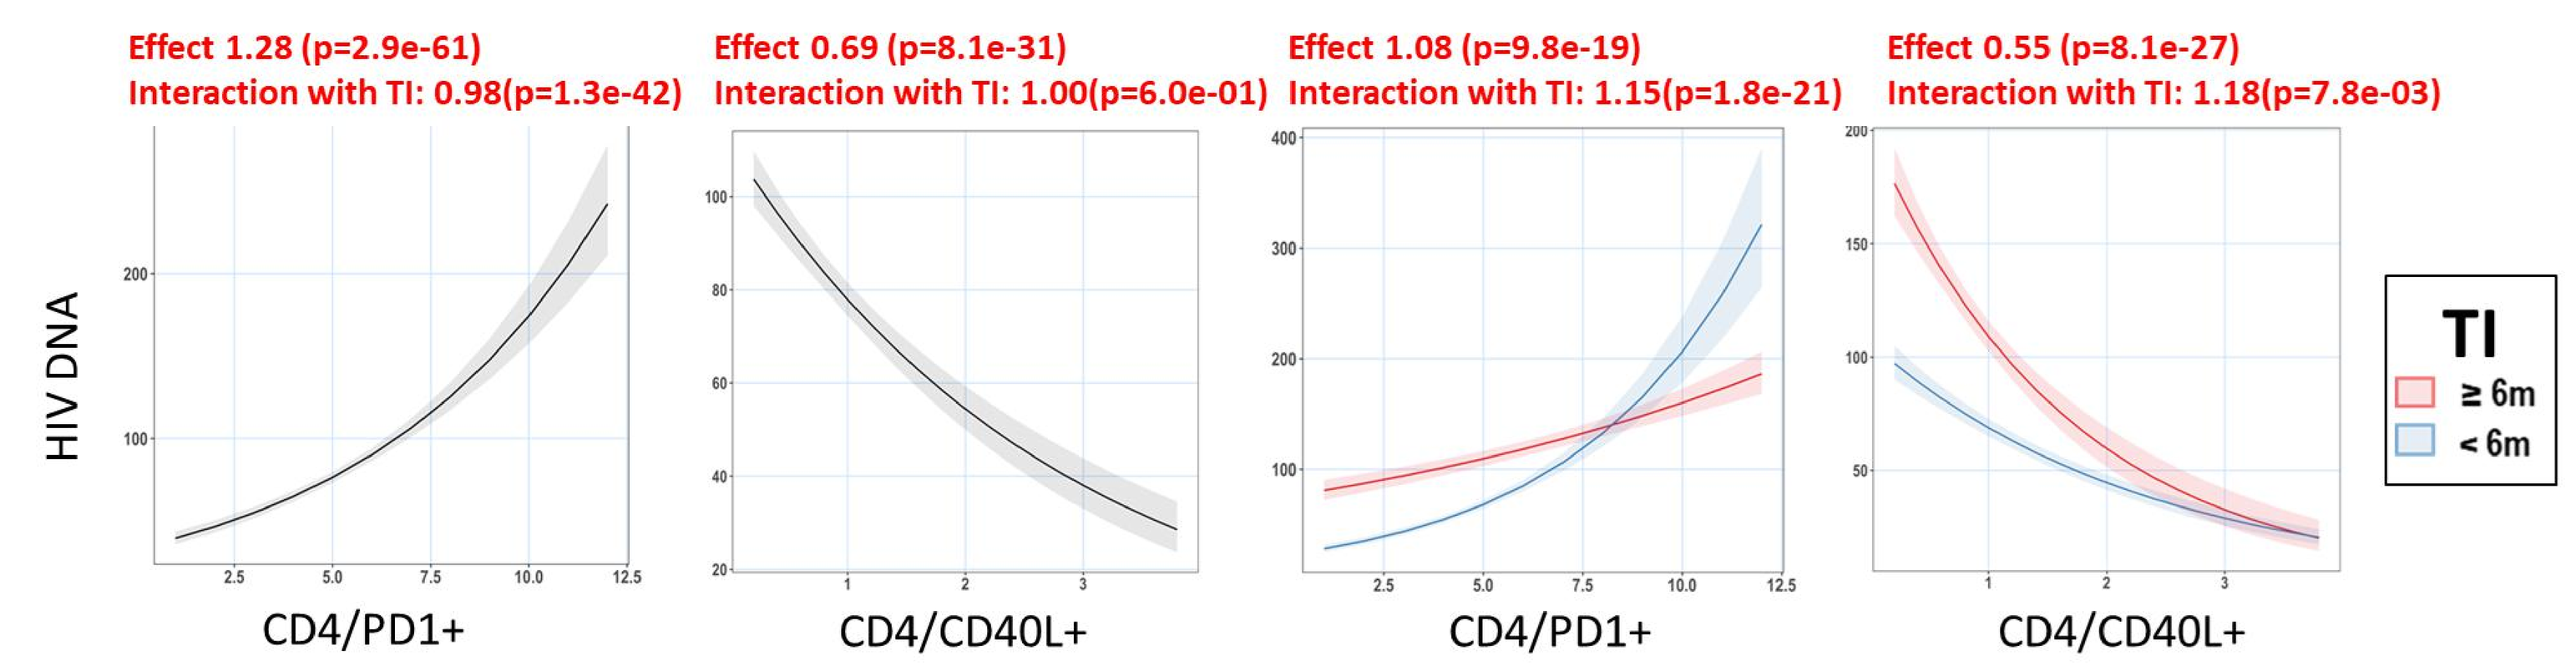
Effect 1.28 (p=2.9e-61)
Interaction with TI: 0.98(p=1.3e-42)
Effect 0.69 (p=8.1e-31)
Interaction with TI: 1.00(p=6.0e-01)
Effect 1.08 (p=9.8e-19)
Interaction with TI: 1.15(p=1.8e-21)
Effect 0.55 (p=8.1e-27)
Interaction with TI: 1.18(p=7.8e-03)
HIV DNA
CD4/PD1+
CD4/CD40L+
CD4/PD1+
CD4/CD40L+
TI
≥ 6m
< 6m

Fig 3. Immunological predictors identified by penalized regression could separate the cohort into 3 distinct clusters

Immunological Predictors	CLUSTER 1 *N=9*	CLUSTER 2 *N=18*	CLUSTER 3 *N=7*	p.overall
CD4+Effector/CD38-HLA-DR+	2.11 [1.27;2.30]	1.60 [0.84;2.51]	1.60 [1.04;2.37]	0.702
CD4+TTM/ICOS+	18.6 [12.3;22.7]	7.69 [6.87;10.0]	16.9 [11.2;18.5]	<0.001
CD4+TTM/ CD38+HLA-DR+	0.54 [0.44;0.75]	0.42 [0.24;0.56]	0.46 [0.35;0.66]	0.260
CD4+Effector/CD25+	28.0 [23.2;30.1]	19.6 [14.2;26.0]	24.9 [22.3;30.3]	0.073
CD4+Effector/TIGIT+	1.51 [1.22;1.71]	1.36 [1.15;1.65]	2.19 [2.02;2.42]	0.006
CD4+/TIGIT+	2.24 [2.18;3.18]	2.37 [2.27;3.00]	4.00 [3.82;4.21]	0.003
CD8+Naïve/TIGIT+	2.29 [1.84;3.25]	2.74 [2.35;3.28]	3.94 [3.54;5.12]	0.011
CD4+/CD40L+	2.28 [2.11;2.54]	0.50 [0.38;1.20]	0.65 [0.45;0.67]	0.001
CD4+/PD1+	2.96 [2.03;3.25]	5.34 [3.27;7.28]	6.52 [5.27;9.61]	0.001

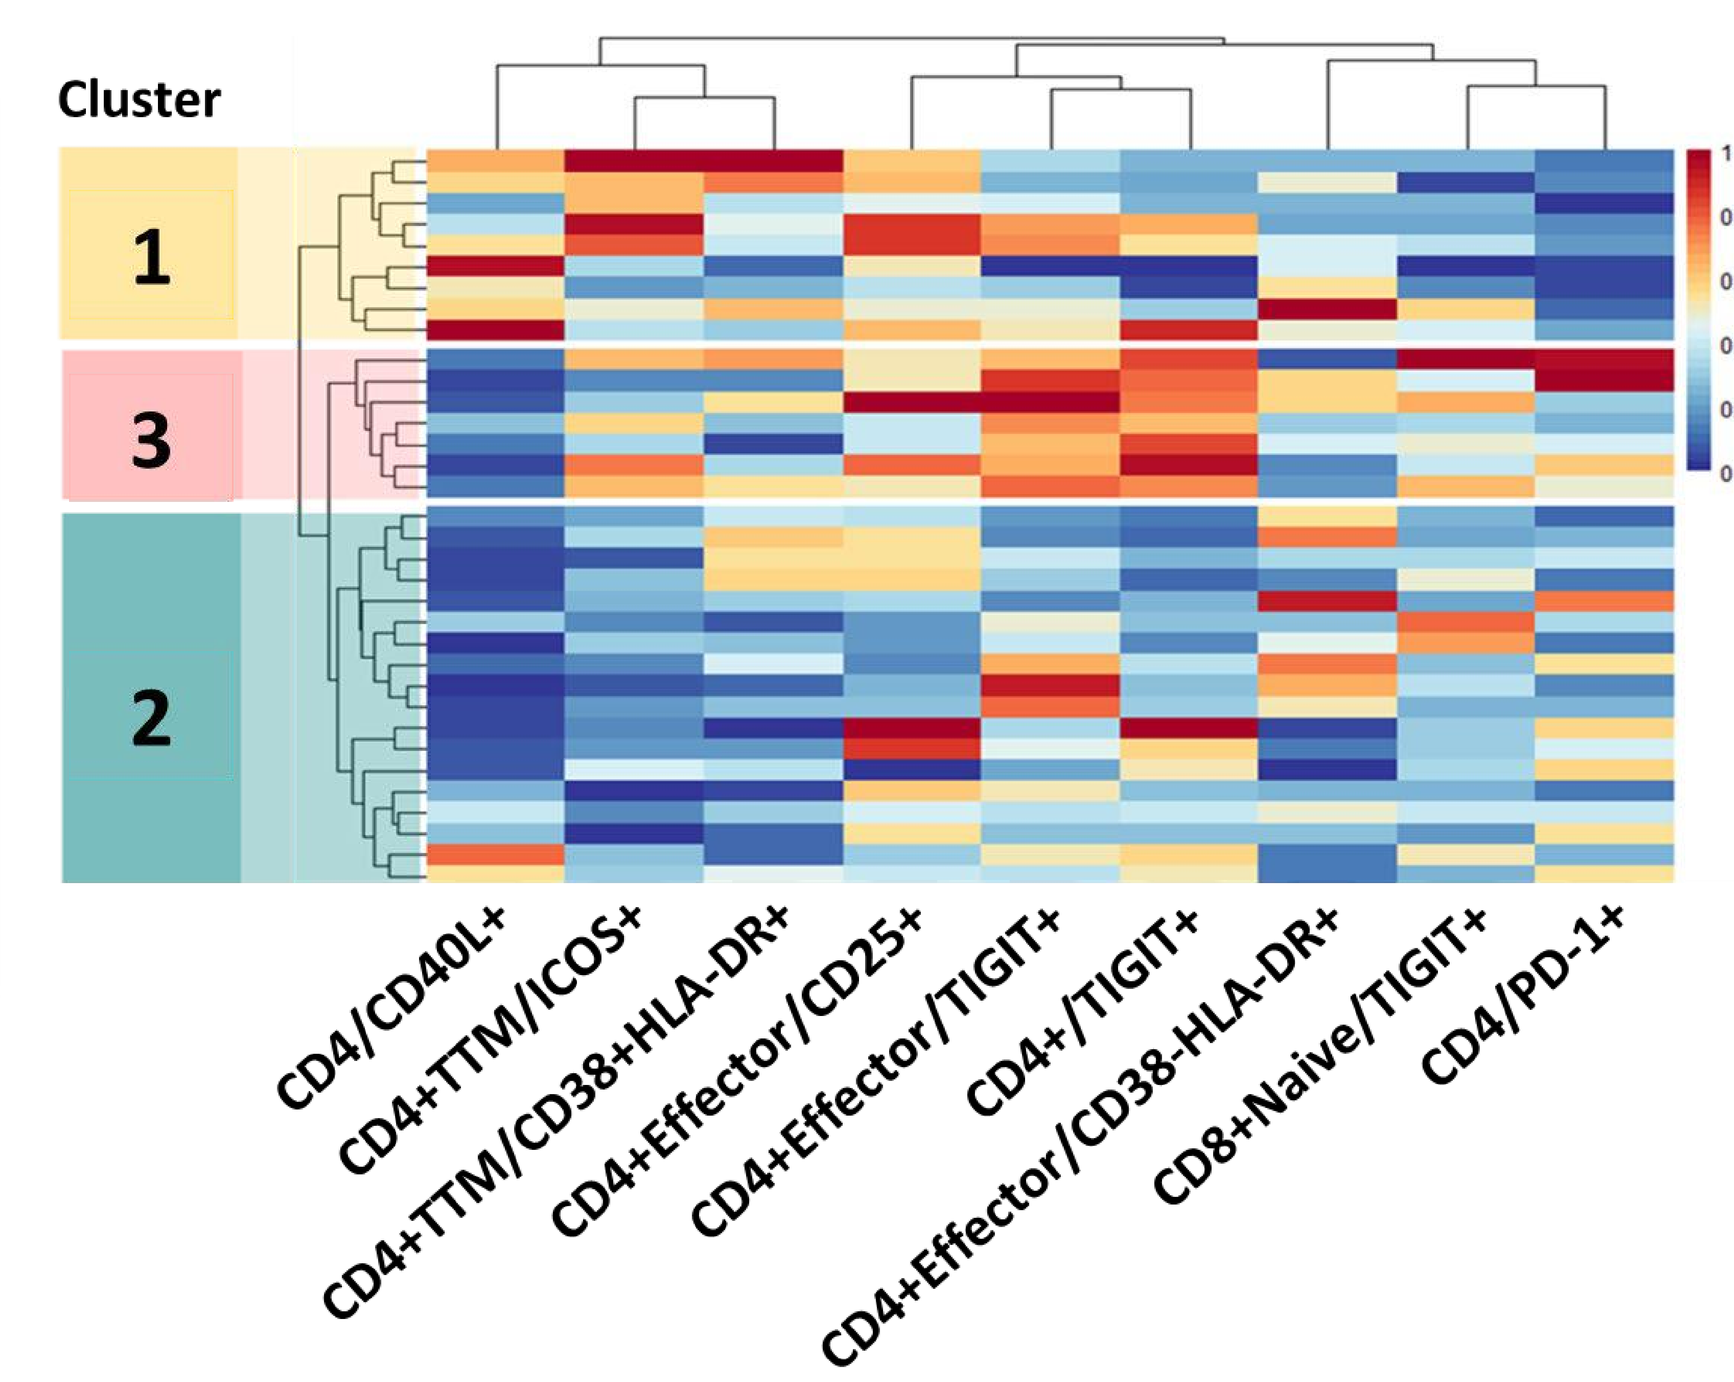
Cluster
1
3
2
CD4/CD40L+
CD4+TTM/ICOS+
CD4+TTM/CD38+HLA-DR+
CD4+Effector/CD25+
CD4+Effector/TIGIT+
CD4+/TIGIT+
CD4+Effector/CD38-HLA-DR+
CD8+Naive/TIGIT+
CD4/PD-1+

Results (contd.)

Fig 4. Clinical differences among the 3 clusters (Fig 3) were identified for TI, cell associated HIV RNA and DNA

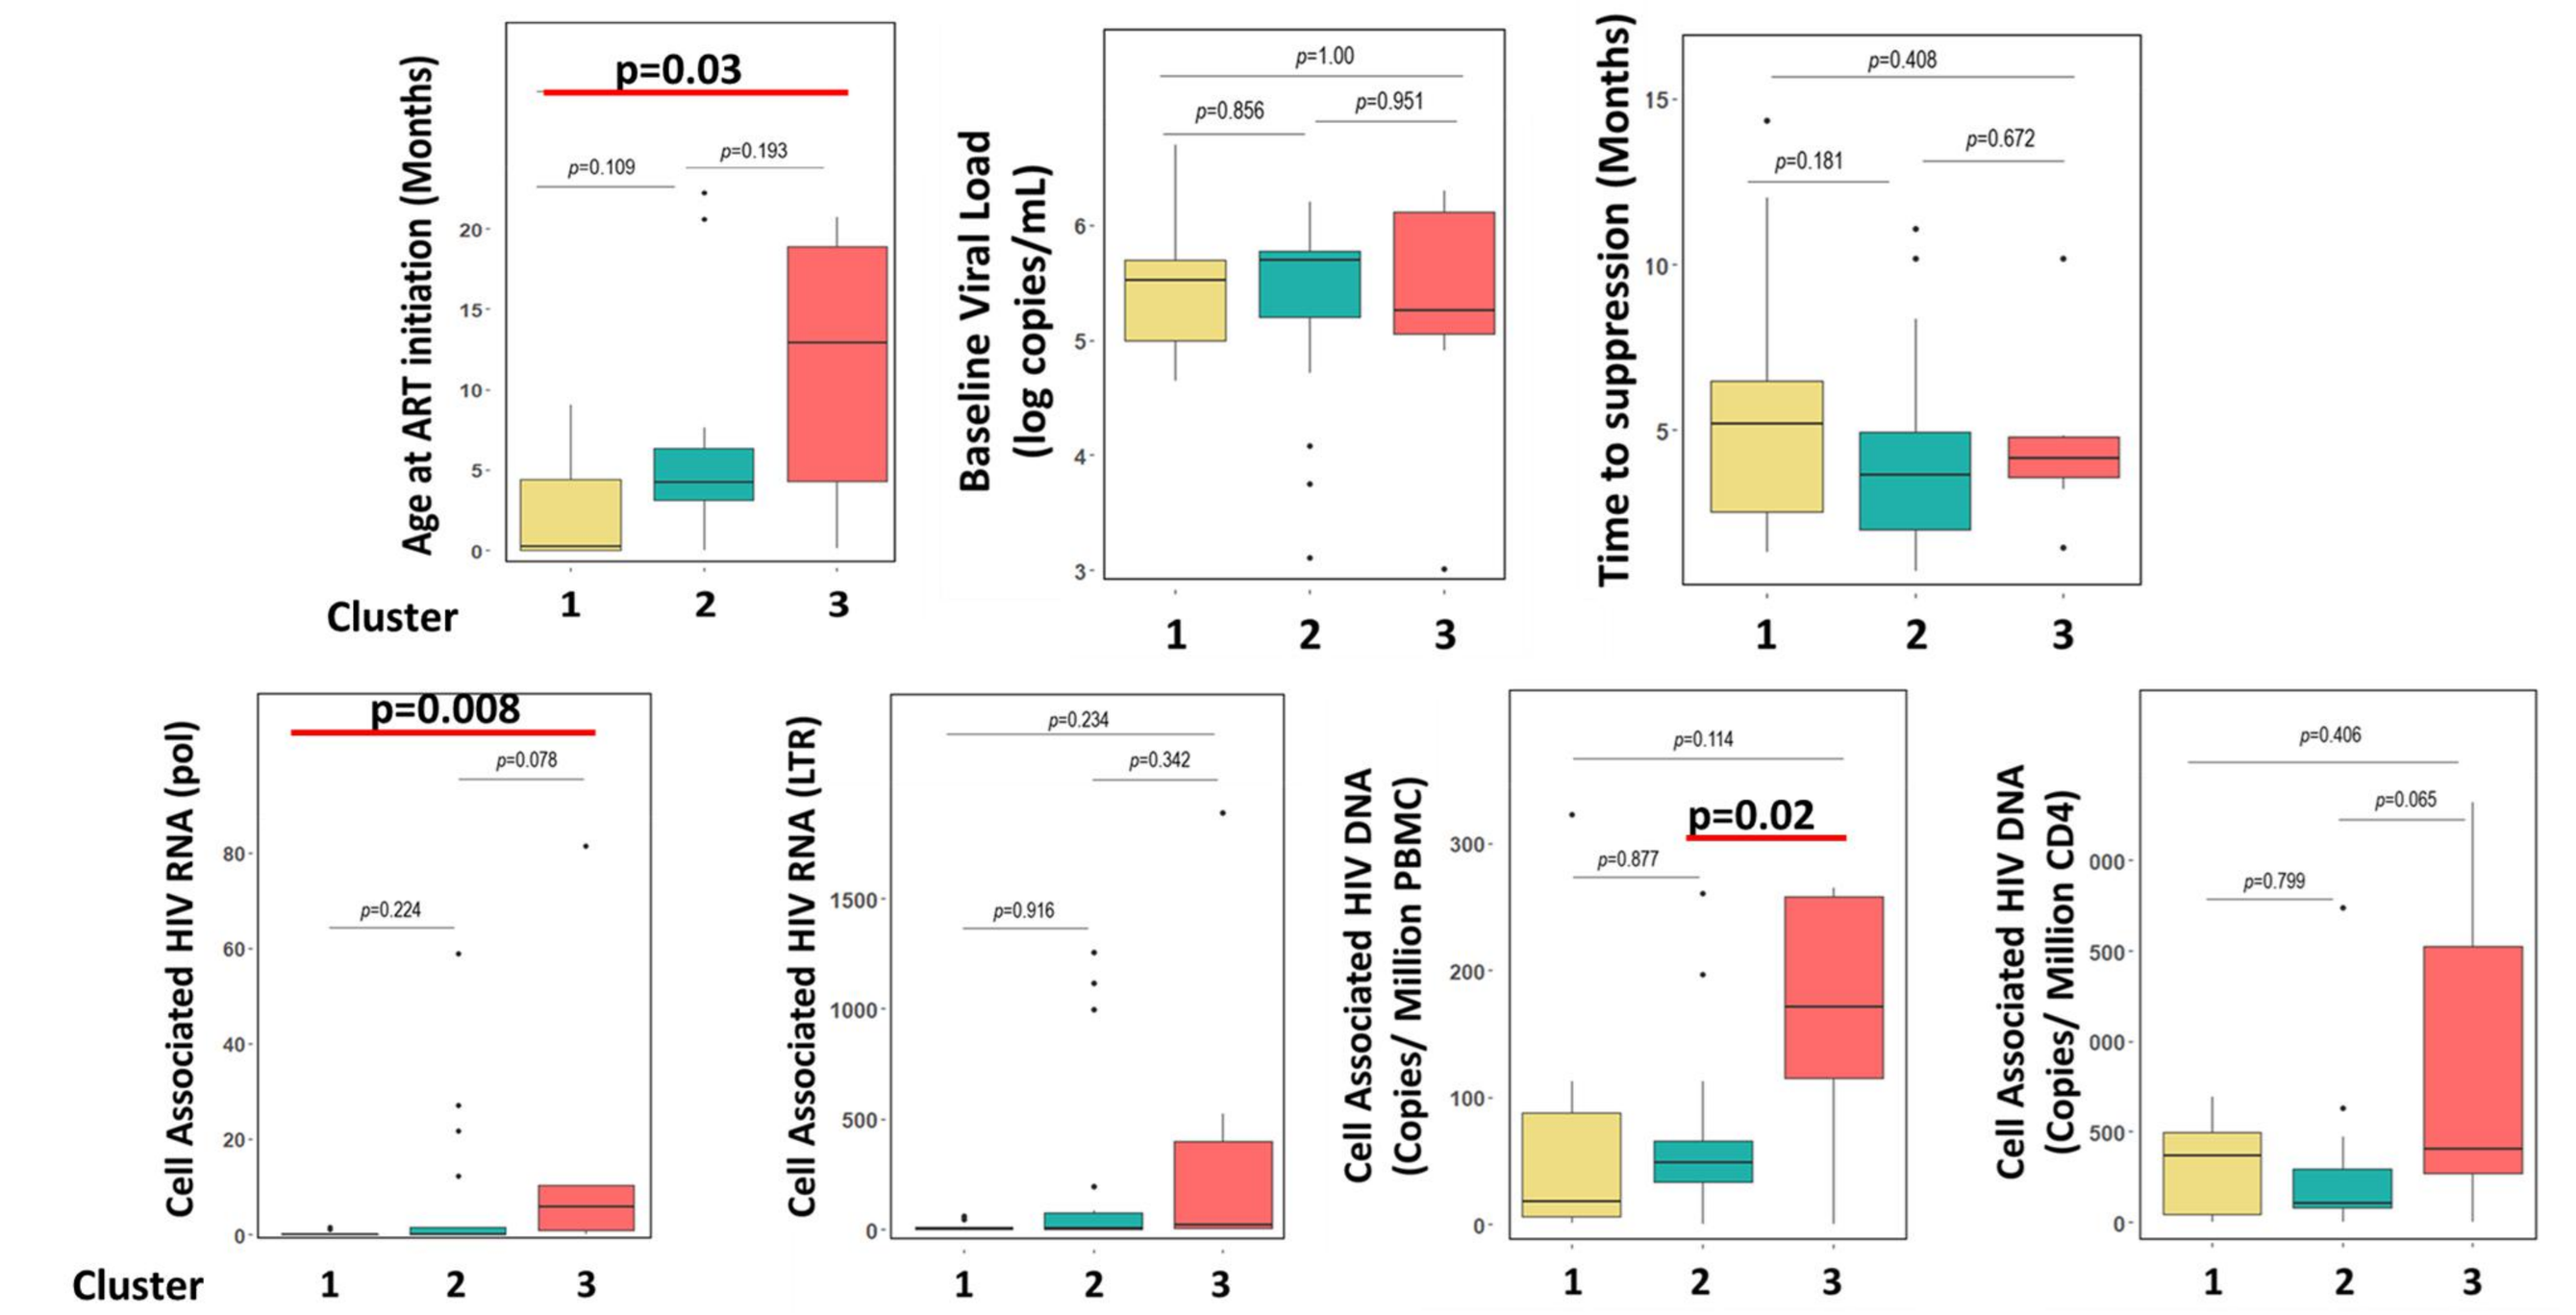
p=0.03
p=0.109
p=0.193
Age at ART initiation (Months)
Cluster
p=1.00
p=0.856
p=0.951
Baseline Viral Load (log copies/mL)
p=0.408
p=0.181
p=0.672
Time to suppression (Months)
p=0.008
p=0.078
p=0.224
Cell Associated HIV RNA (pol)
p=0.234
p=0.342
p=0.916
Cell Associated HIV RNA (LTR)
p=0.114
p=0.02
p=0.877
Cell Associated HIV DNA (Copies/ Million PBMC)
p=0.406
p=0.065
p=0.799
Cell Associated HIV DNA (Copies/ Million CD4)

Fig 5. CD4 and CD8 T cell functions were negatively associated with baseline frequency of PD-1+ CD4 T cells

PD-1 expression on CD4 T cells negatively associates with:

1) proportion of antigen responsive CD4 and Tfh cells

2) CD8 T cells expressing the degranulation marker CD107a

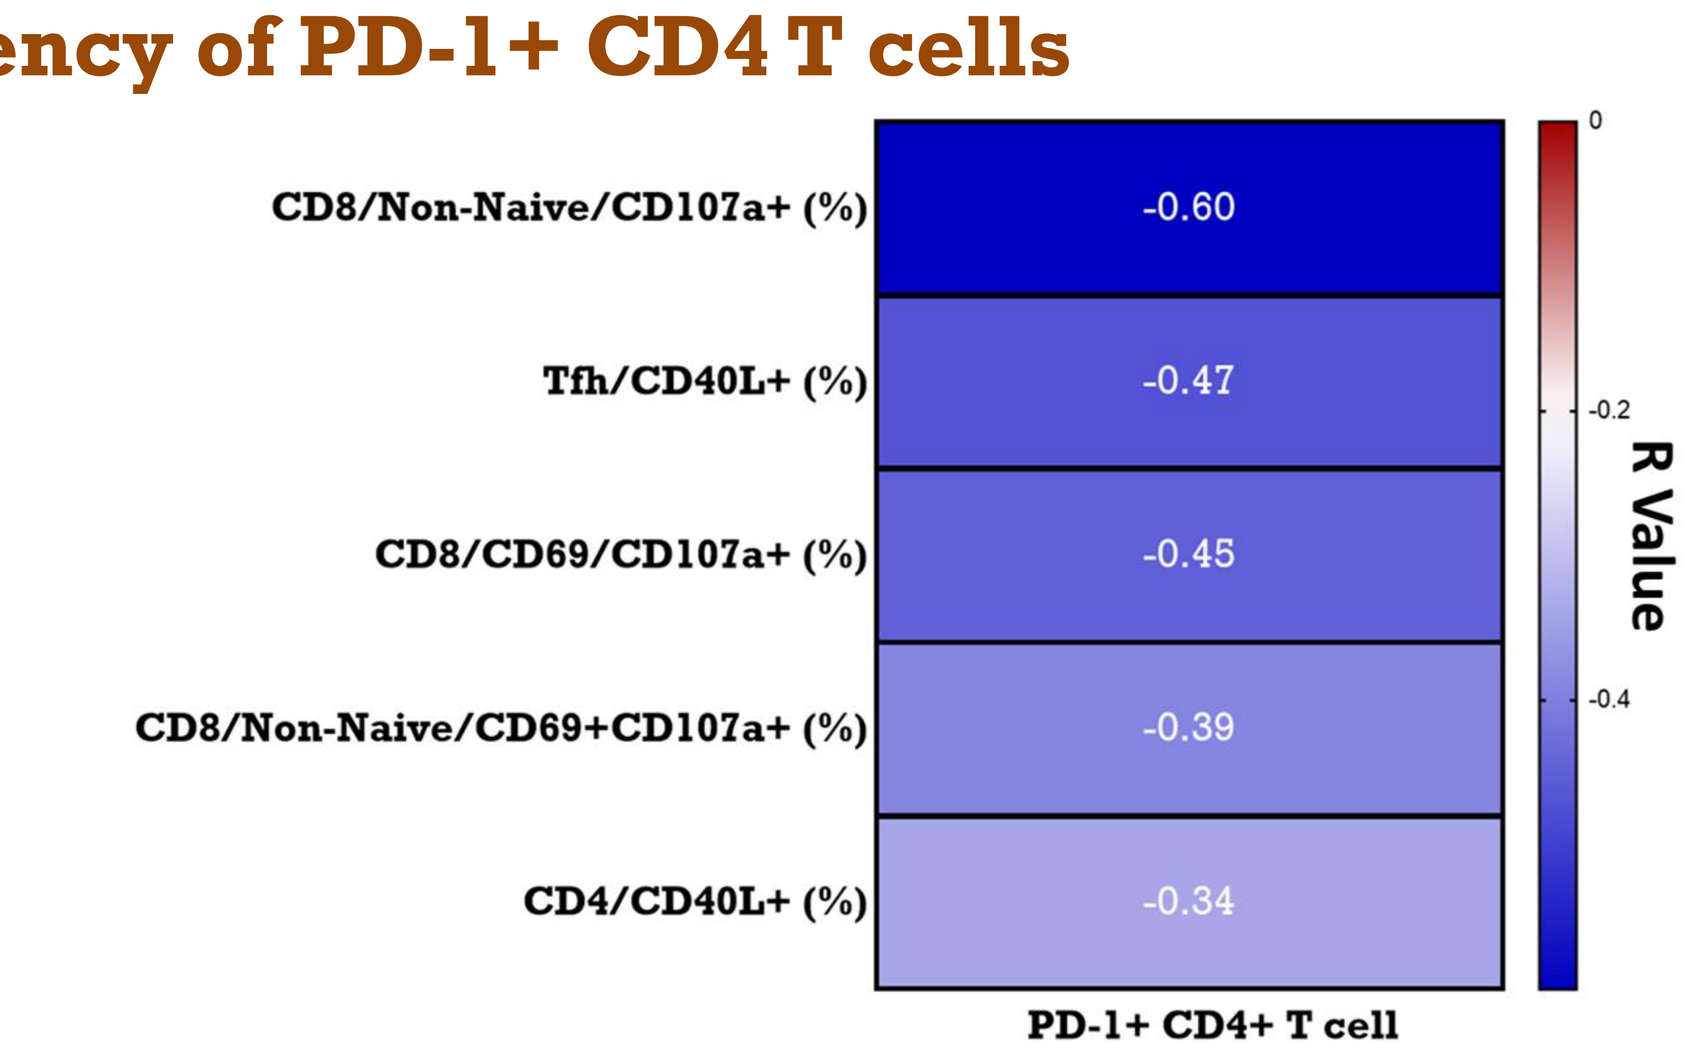
CD8/Non-Naive/CD107a+ (%)
-0.60
Tfh/CD40L+ (%)
-0.47
CD8/CD69/CD107a+ (%)
-0.45
CD8/Non-Naive/CD69+CD107a+ (%)
-0.39
CD4/CD40L+ (%)
-0.34
PD-1+ CD4+ T cell
R Value

Summary and Conclusions

PD1 expression on CD4 T cells and GAGsp CD4 T cells functionality discriminate between participants with high and low HIV-DNA

Cluster 1	Cluster 2	Cluster 3
Low reservoir size Early Treated High % CD4+TTM/ICOS+ Low % of CD4/TIGIT+ High % of CD4/CD40L+ Low % of CD4/PD-1+	Medium reservoir size Low% CD4+TTM/ICOS+ Low% CD4+Effector/TIGIT+ Lower % of CD4/CD40L+	High reservoir size Late treated Low% CD4+TTM/ICOS+ High%CD4+Effector/TIGIT+ High % CD4/TIGIT+ High % of CD4/PD1+

Proposed model: PD-1+ CD4 T cells are associated with impaired GAGsp CD4 and CD8 T cell functions

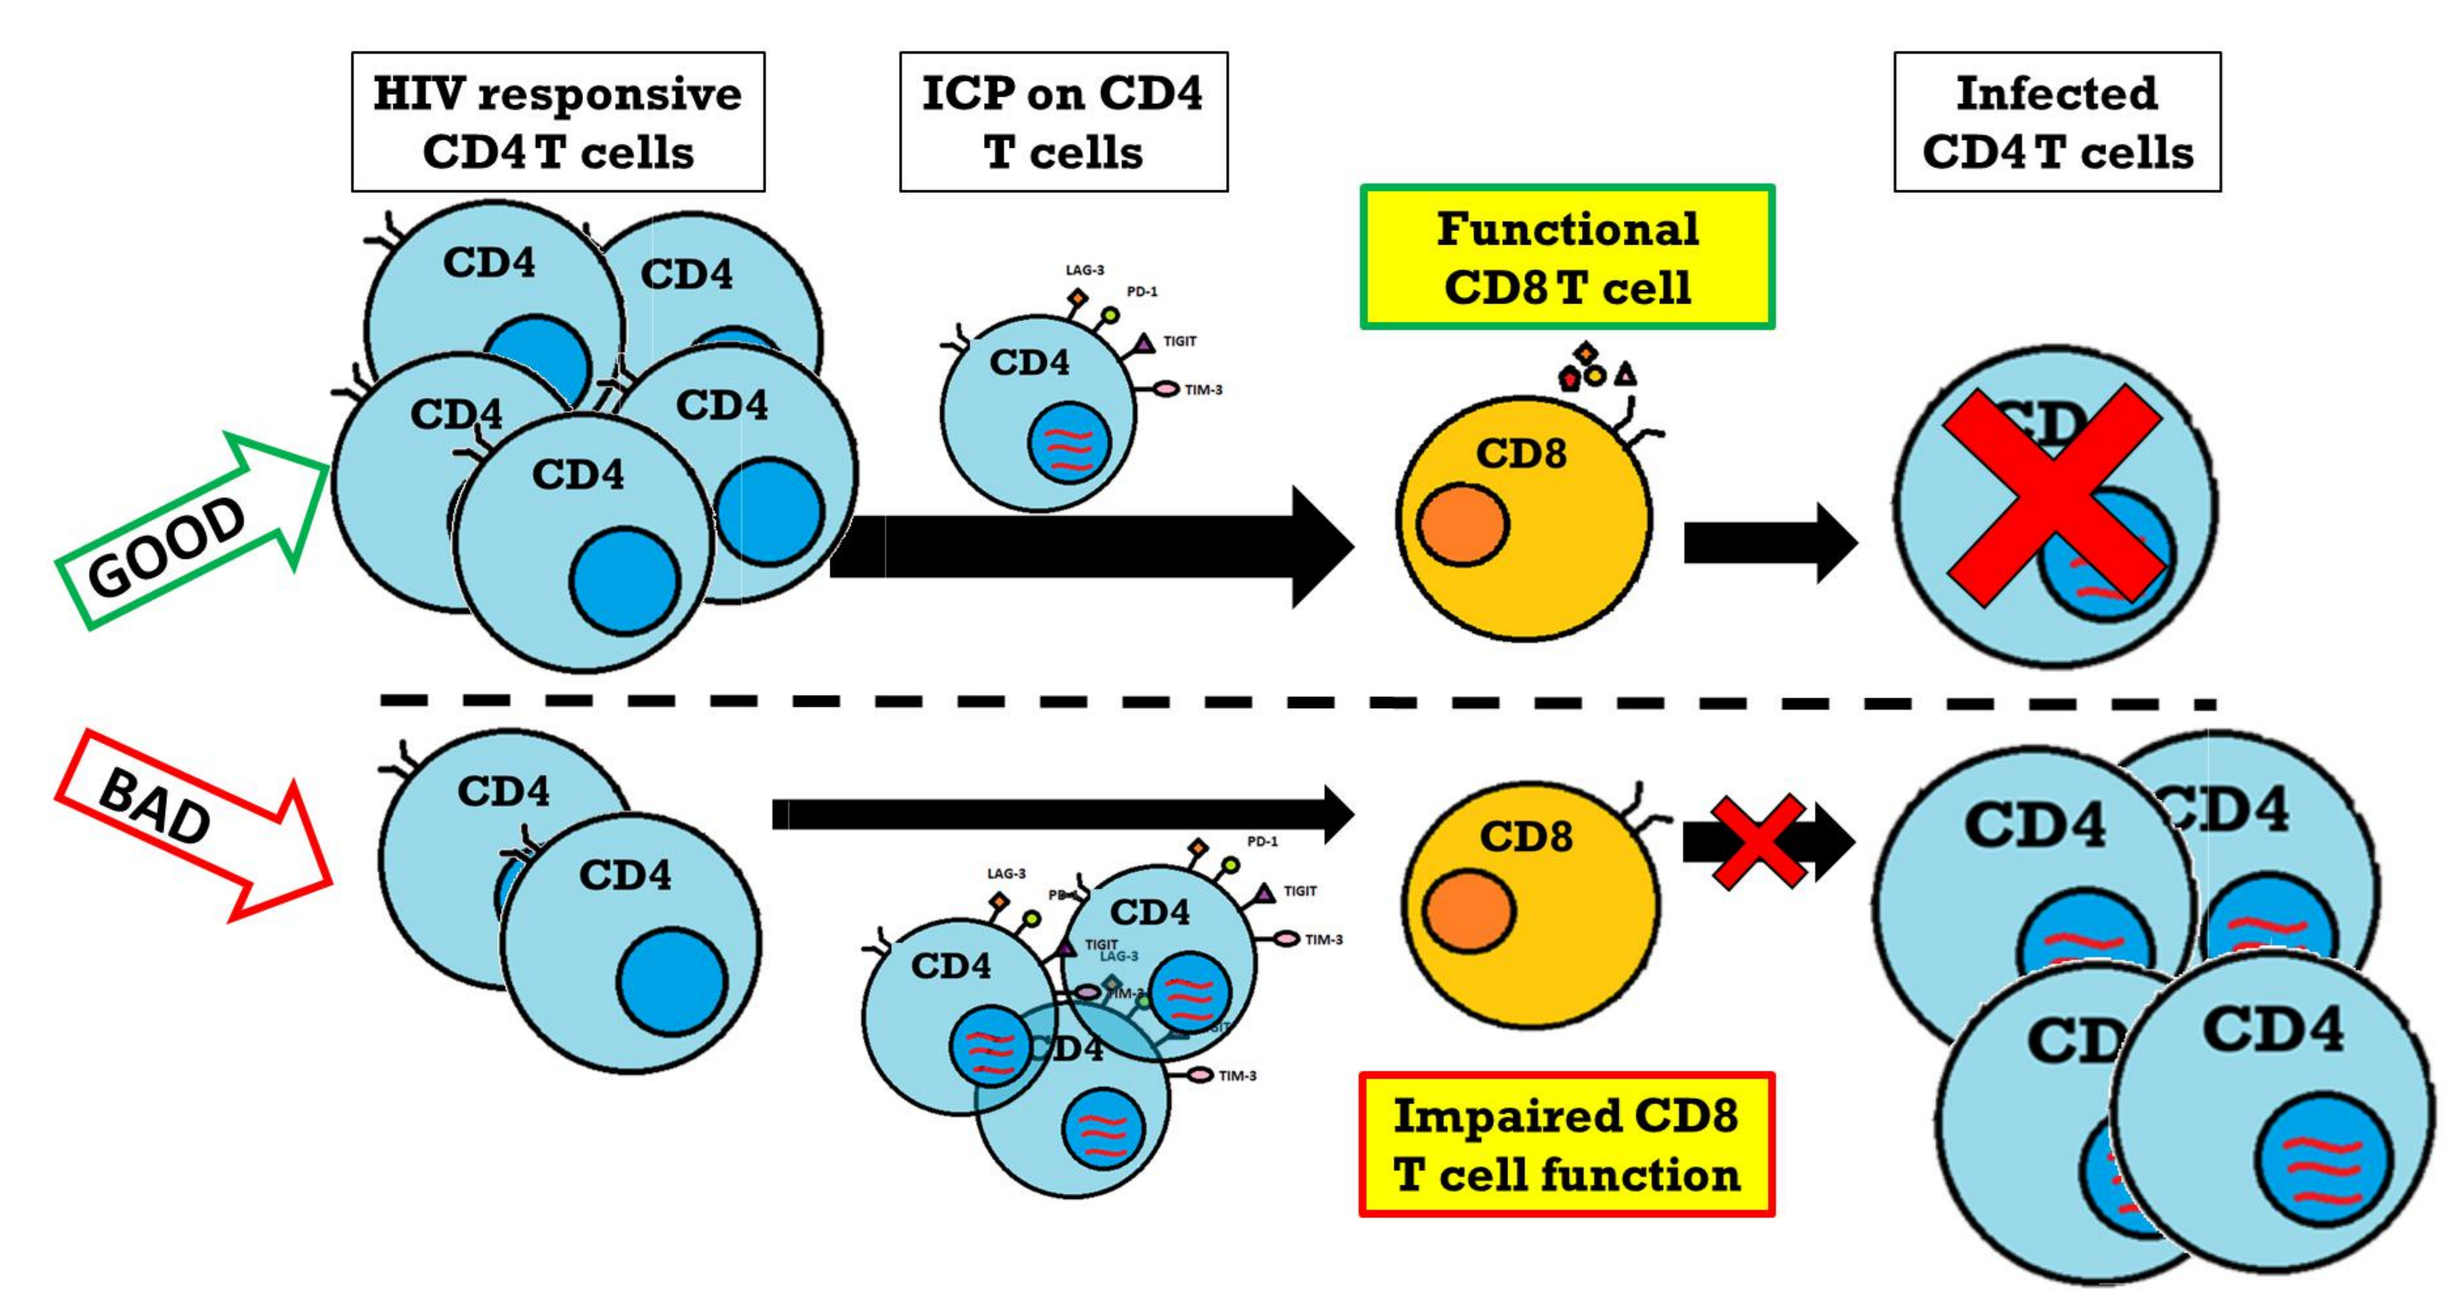
HIV responsive CD4 T cells
ICP on CD4 T cells
Functional CD8 T cell
Infected CD4 T cells
GOOD
BAD
CD4
CD8
Impaired CD8 T cell function

Acknowledgements

This work was supported by the EPIICAL project (http://www.epiical.org/), supported by PENTA-ID Foundation (http://penta-id.org/), funded through an independent grant by ViiV Healthcare UK. . We thank the Laboratory Sciences Core of the Miami Center for AIDS Research (P30AI073961) for technical support. We thank Flow Cytometry Core Facility of the University of Miami for instrumentation and study participants for consenting to this project.